Canine Splenic Hemangiosarcoma treated by Autologous Monocyte-Derived Dendritic Cell Therapy

[a]Spiller, V., [b]Vetter, M., [c]Grammel, T.

[a]PetBioCell GmbH, Osterode am Harz, Germany, [b]Department of Gynaecology, Martin Luther University Halle-Wittenberg, Halle (Saale), Germany, [c]Tiergesundheitszetrum Suedharz, Osterode am Harz, Germany

Background and Objective

Canine hemangiosarcoma is known for progressive course of disease and short survival time between 17 - 39 days at given palliative treatment[1, 2, 3], only. Amongst various therapy protocols such as irradiation or doxorubicin-based chemotherapy treatment, cellular immunotherapy based on sole use of autologous dendritic cells (DCs) or DCs administered after propranolol pre-treatment of the patient are considered valuable options to attain a similar rate of survival time but with undisturbed animal health in favor of animal welfare and dog owner's joy. For both scenarios, our recent studies are presented.

[1]Fidel, J., Schiller, I., Hauser, B., et al. Histiocytic sarcomas in flat-coated retrievers: a summary of 37 cases (November 1998 - March 2005). Vet. Comp. Oncol. 2006; 4:63-74. doi:10.1111/J.1476-5810.2006.00090.X
[2]Dervisis, N., Kuipel, M., Qin, Q., et al. Clinical prognostic factors in canine histiocytic sarcoma. Vet.Comp. Oncol. 2016; 1-10. doi:10.1111/vco.12252
[3]Skorupski, K., Rodriguez, C., Krick, E., et al. Long-term survival in dogs with localized histiozytic sarcoma treated with CCNU as an adjuvant to local therapy. Vet. Comp. Oncol. 2009; 7: 139139-144. doi:10ß.1111/j.1476-5829.2009.00186.x

Standard Immunotherapy - Dendritic Cell Vaccine

For a prospective study[4], dogs (n=452) were diagnosed with splenic hemangiosarcoma and splenectomized. Of these, 42 dogs with hemangiosarcoma stage II were subjected to sole DC therapy and monitored during the period of Jan 1, 2016 – Dec 31, 2022 (Fig. 1 and Fig. 2).

Of each dog, autologous monocyte-derived DCs were propagated in RPMI 1640 medium supplemented with canine GM-CSF and canine IL-4 at GMP-compliant evironment for seven days. For dogs DCs-treated at least on three occasions (n=34), cells were resuspended in isotonic sodium chloride solution (0.9 %) at quantities ranging between 4.35 – 8.17 x 10^6 at 95% CI of which at least 10 % of cells expressed the CD-1b surface molecule. Each cell suspension was administered intradermally with 24 hours.

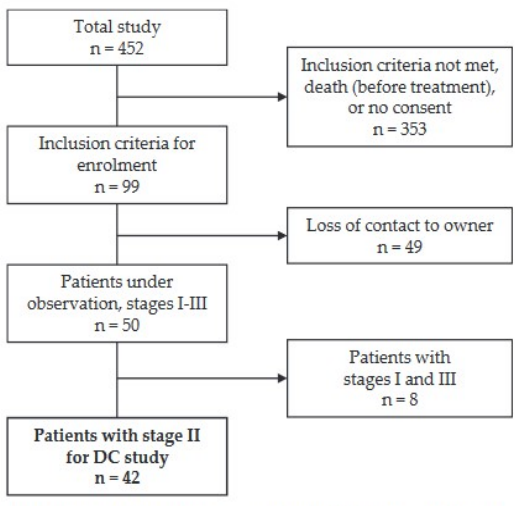
Total study
n = 452
Inclusion criteria not met, death (before treatment), or no consent
n = 353
Inclusion criteria for enrolment
n = 99
Loss of contact to owner
n = 49
Patients under observation, stages I-III
n = 50
Patients with stages I and III
n = 8
Patients with stage II for DC study
n = 42

Fig. 1. Consort diagram illustrating critical selection procedure of 42 candidate patients fit for study purpose. Abbreviations: DC: dendritic cell.

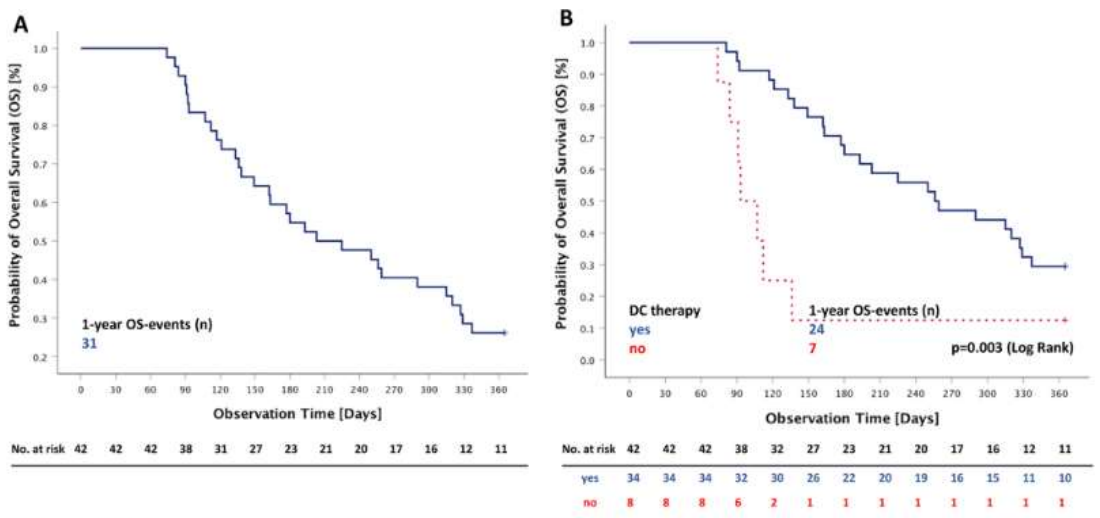
A
Probability of Overall Survival (OS) [%]
1-year OS-events (n)
31
Observation Time [Days]
No. at risk 42 42 42 38 31 27 23 21 20 17 16 12 11
B
Probability of Overall Survival (OS) [%]
DC therapy
yes
no
1-year OS-events (n)
24
7
p=0.003 (Log Rank)
Observation Time [Days]
No. at risk 42 42 42 38 32 27 23 21 20 17 16 12 11
yes 34 34 34 32 30 26 22 20 19 16 15 11 10
no 8 8 8 6 2 1 1 1 1 1 1 1 1

Fig. 2. Survival estimates of dogs diagnosed with stage II hemangiosarcoma of the spleen. Upon splenectomy, dogs were subjected to dendritic cell therapy. A: Overall survival probability of the total DC therapy study. B: Overall survival probability considering dendritic cell therapy. Abbreviations: DC: dendritic cell.

The study results revealed an overall median survival time of 203 days including all 42 dogs, and 256 days for those receiving at least three DCs vaccines (n=34). Of these, 10 dogs (29 %) demonstrated a survival rate of one year.

[4]Spiller, V., Vetter, M., Dettmer-Richardt, C., Grammel, T. Prospective study of successful autologous dendritic cell therapy in dogs with splenic stage II hemangiosarcoma. The Veterinary Journal, , DOI: 10.1016/j.tvjl.2024.106196

Modified Immunotherapy - Propranolol and Dendritic Cell Vaccine

In November 2021, propranolol-based immunotherapy of splenic hemangiosarcoma was reported by Dr. Erin Dickerson at the virtual VCS Conference 2021[5]. Further inspired by the recent publication of Ammons et al. (2023)[6] we elaborated on the protocol by administration of propranolol immediately upon diagnosis (day 0) followed by five treatments at varying intervals of autologous dendritic cell suspension during a period from day 14 to day 180 (Tab. 1) after disease diagnosed. The case study refers to a male Staffordshire Bullterrier, aged 13 years old and weighing 16.4 kg.

Table 1. Scheme of propranolol treatment in combination with DCs-based immunotherapy.

Days after Diagnosis	Mode of Treatment
0	propranolol, 1 mg/kg body weight, daily
	ultrasound control
14	dendritic cell vaccine
40	dendritic cell vaccine
60	dendritic cell vaccine
97	ultrasound control
120	dendritic cell vaccine
180	dendritic cell vaccine
384	ultrasound control

[5]Dickinson, E. 2021.Using Propranolol to treat canine hemangiosarcoma:Lesson learned and new directions. **Course:** 2021 VCS Annual Conference On-Demand (Hemanigosarcoma Session).
[6]Ammons DT, MacDonald CR, Chow L, Repasky EA, Dow S. Chronic adrenergic stress and generation of myeloid-derived suppressor cells: Implications for cancer immunotherapy in dogs. *Vet Comp Oncol*. 2023; 21(2): 159-165. doi:10.1111/vco.12891

On February 23, 2023, a volume of 300 ml of hemosuccus was found in the abdominal cavity. In addition, a fist-sized tumor of spleen and metastatic signs in the liver tissue (Fig. 3) were identified. The splenectomy was done immediately. The clinical staging classified a splenal hemangiosarcoma stage III (T1, M1).

Propranolol is a medication of the beta-blocker class and dosed 1 mg/kg of body weight. As summarised in Table 1, it was given the day when hemangiosarcama was diagnosed (day 0) followed by administration of autologous monocyte-derived dendritic cells on five occasions (March 2023 - August 2023). Daily propranolol treatment continued without observation of any serious clinical signs. Since diagnosed and treatment initiated, the dog has fully recovered and keeps alive in good general health (October 2024).

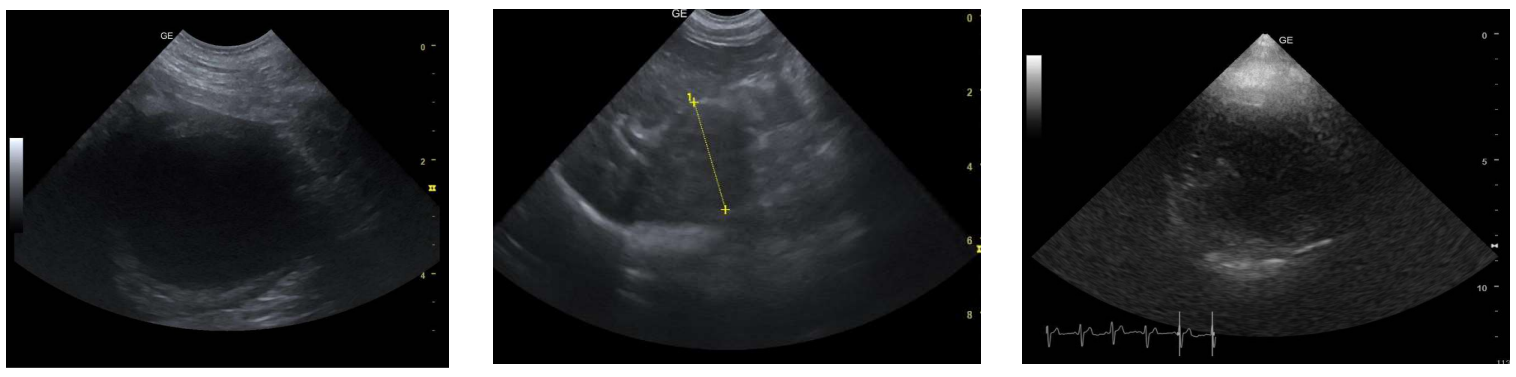

Fig. 3. The medical records refers to three ultrasound images visualizing changes of the liver appearance. The left-sided image was performed in emergency mode shortly before splenectomy (February 23, 2023). The surgical report mentions several metastatic changes in the liver. The centered control image was taken on May 31, 2023. On June 17, 2024, a cardiac ultrasound examination was performed to check the liver parenchyma (right-sided image). Overall, regeneration/sclerosis of the old tumor area kept increasing over time.

Conclusion

The sole use of autologous DCs in canine splenic hemangiosarcoma demonstrated promising survival times in favor of animal welfare and owner's joy. Moreover, there is some evidence that pre-treatment of propranolol prior to application of DCs could even further improve the patient's life expectancy. The authors are currently elaborating on the subject including larger number of dogs for study purpose.

Orlando, Fl
Oct 16 – 19, 2024